IDENTIFICATION AND CHARACTERIZATION OF A NEW CYTOTOXIC AGENT FROM ACTINOMYCETES (ECO-04601)

V. Dimitriadou[1], J. McAlpine[1], E. Zazopoulos[1], B. Bachman[1], F. Berger[2], L. Pelletier[2], M. Alaoui-Jamali[3], C. Farnet[1]

[1] Ecopia BioSciences Inc., Saint-Laurent, QC, Canada [2] Laboratoire de Neurobiologie Préclinique, INSERM U318, Grenoble, France [3] Lady Davis Institute, Montreal, QC, Canada

2060

INTRODUCTION

ECO-04601 structure: farnesylated dibenzodiazepinone

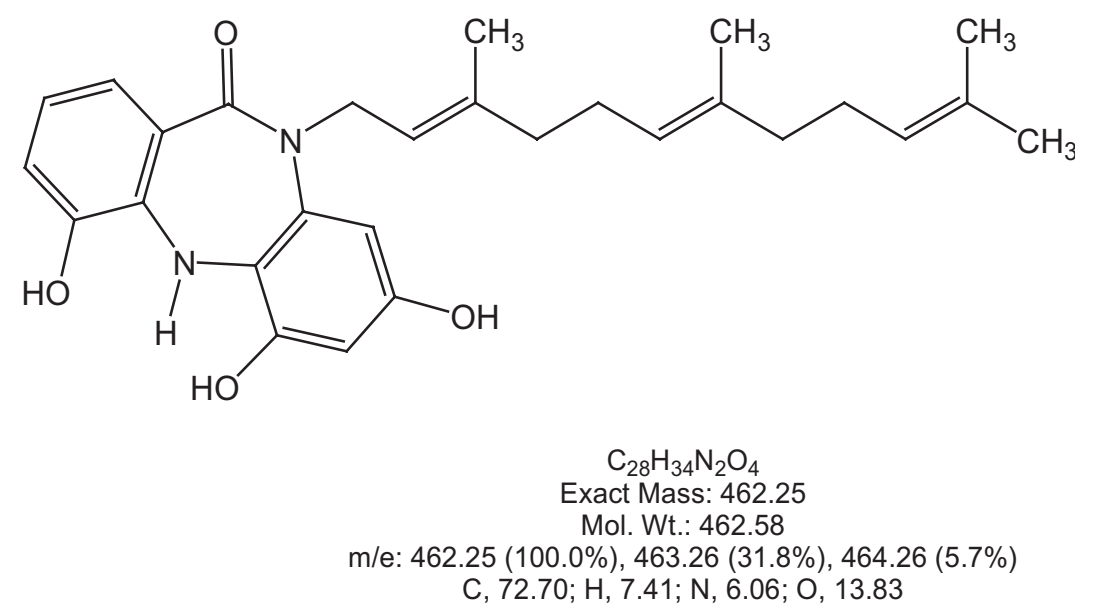
O
CH3
CH3
CH3
CH3
N
N
H
HO
OH
HO
C28H34N2O4
Exact Mass: 462.25
Mol. Wt.: 462.58
m/e: 462.25 (100.0%), 463.26 (31.8%), 464.26 (5.7%)
C, 72.70; H, 7.41; N, 6.06; O, 13.83

ECO-04601 is a structurally novel compound discovered using Ecopia's genomic platform (Zazopoulos et al., Nat. Biotechnol. 21(2) 187-190, 2003) through analysis of over 500 actinomycete loci encoding bioactive compounds. ECO-04601 was expressed in the fermentation broth of an actinomycete (*Micromonospora* sp.) and purified by a series of chromatographic methods. The compound is virtually neutral and very lipophilic.

BACKGROUND

Ecopia's discovery platform

Ecopia's innovative DECIPHER® technology platform revolves around the discovery of lead compounds that have been optimized through years of natural evolution in bacteria. The DECIPHER® technology uses a combination of leading edge genomics and bioinformatics to make computer predictions of the chemical structure of potential new drugs based on gene sequence information obtained by scanning the bacterial genome. For more information please visit Ecopia's website at www.ecopiabio.com.

Scientific rationale for developing ECO-04601 as an anticancer agent

This novel molecule has shown anti-proliferative activity against several human cancer cell lines from the NCI panel. The results show that ECO-04601 in the µM concentration range inhibits the growth of leukemia cells and cancer cell lines derived from lung, colon, brain, ovary, renal, prostate and breast tissues as well as melanomas.

RESULTS

Effect of ECO-04601 against gliomas

In vitro results:

Table 1: Effect of ECO-04601on tumor cell growth

TUMOR TYPE	ORIGIN	CELL LINES	IC_{50} (µM) MEAN	SD	n
Oligodendrogliomas	HUMAN	GHA	1.6	0.70	10
	HUMAN	DN	2.9	0.72	4
Astrocytomas	HUMAN	U373	3.8	1.37	4
	HUMAN	GHD	6.5	2.86	8
Glioblastomas	HUMAN	GL26	8.9	1.10	4
	RAT	C6	4.3	2.29	5
Gliosarcoma	RAT	9L	8.3	3.78	4

- Plated cells were incubated for 96 hrs with 6 concentrations (0.001 to 10 µM) of ECO-04601 diluted in 0.5% DMSO/culture medium. Effect on cell proliferation was measured using the MTT proliferation assay.
- Results are expressed as the mean ± standard deviation of the IC50 obtained from 4 to 10 independent experiments (in sixplicate wells) and calculated using the GraphPad software.

Data show that low µM concentrations of ECO-04601 induce inhibition of glioma cell proliferation independently of their origin.

In vivo results:

Figure 1: Inhibition of growth of established glioma tumors by ECO-04601

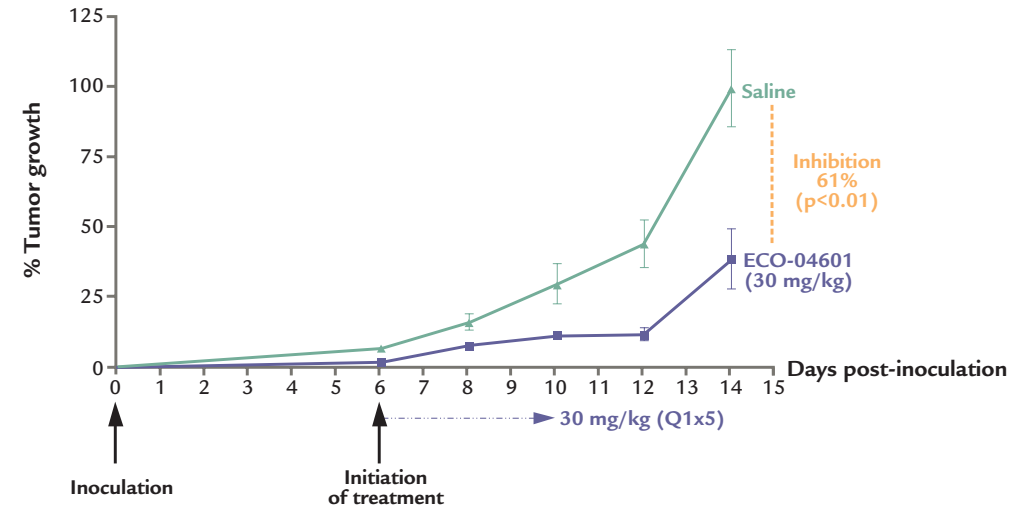
% Tumor growth
Saline
Inhibition 61% (p<0.01)
ECO-04601 (30 mg/kg)
Days post-inoculation
30 mg/kg (Q1x5)
Inoculation
Initiation of treatment

Figure 2: Dose-response effect of ECO-04601 on growth of established glioma tumors

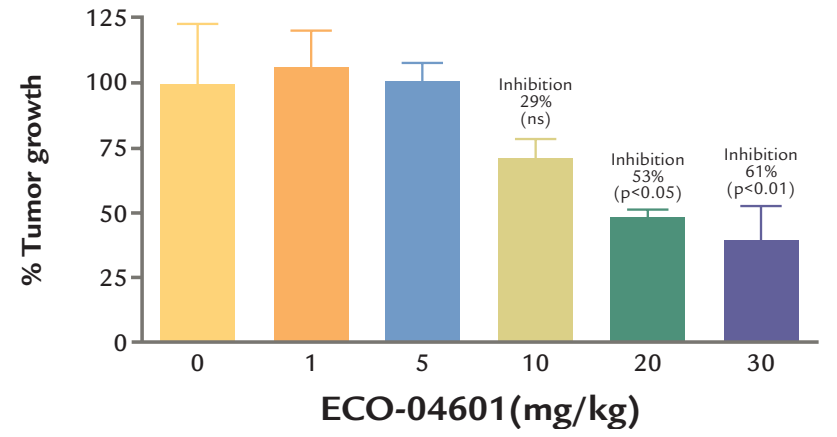
% Tumor growth
Inhibition 29% (ns)
Inhibition 53% (p<0.05)
Inhibition 61% (p<0.01)
ECO-04601(mg/kg)

- The effect of ECO-04601 was evaluated on nude mice bearing subcutaneous tumors from rat C6 glioma cells. Mice were treated once daily i.p. for 5-7 consecutive days.
- Statistical analysis (Anova's one-way analysis of variance) of the mean values of the tumor volume from saline- and ECO-04601-treated groups revealed a dose-dependent inhibition of tumor growth by ECO-04601.

Doses of 20 and 30 mg/kg induced statistically significant inhibition of tumor growth as compared to the saline-treated group.

Figure 3: Effect of ECO-04601 treatment on the morphology of glioma C6 tumors

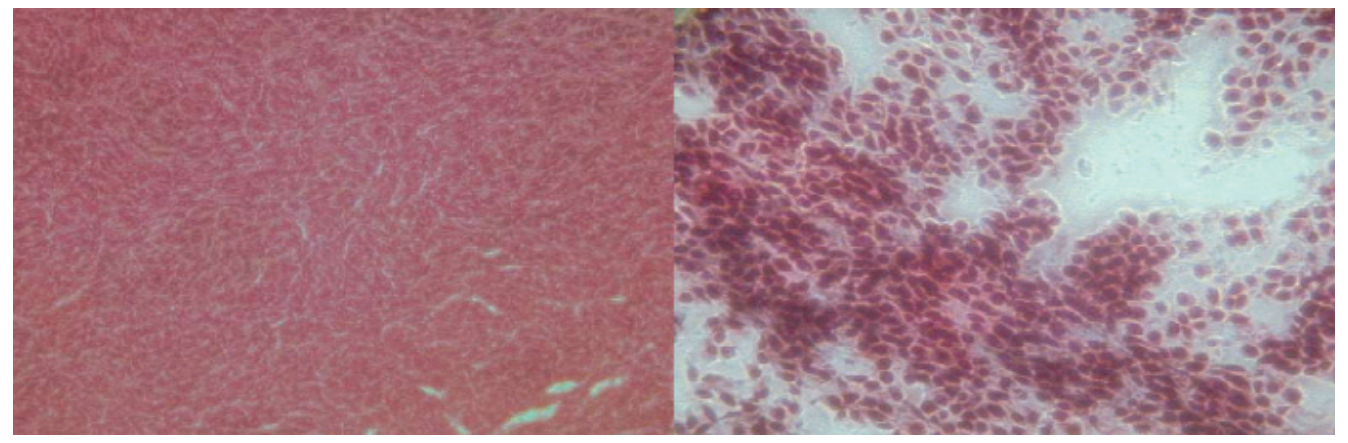

Saline ECO-04601 (20 mg/kg)

Tumors from saline- and ECO-04601-treated mice (established tumor model) were removed after sacrifice and prepared for histological examination. Cryostat sections (10 µm) were fixed in buffered formalin 10% and stained with Masson's trichrome dye. Representative pictures of tumor sections from control and treated animals are shown (x20).

Microscopic examination revealed a significant decrease of tumor cell density in ECO-04601-treated tumors with nuclei in these cells appearing larger than in cells of untreated tumors.

Figure 4: Inhibition of growth of newly implanted tumors by ECO-04601

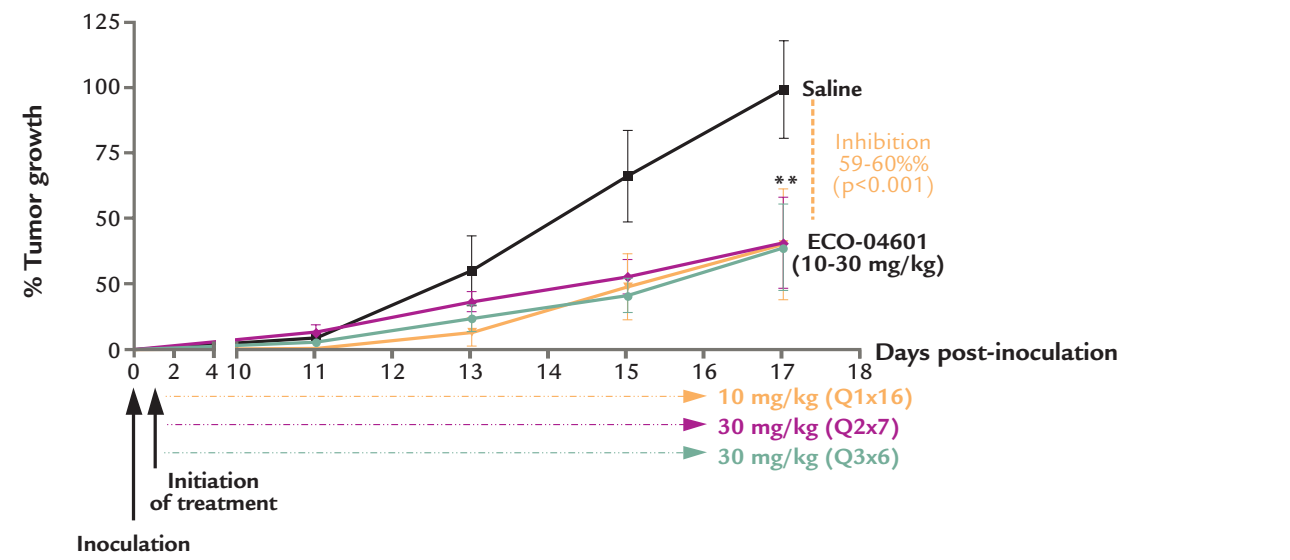
% Tumor growth
Saline
Inhibition 59-60%% (p<0.001)
ECO-04601 (10-30 mg/kg)
Days post-inoculation
10 mg/kg (Q1x16)
30 mg/kg (Q2x7)
30 mg/kg (Q3x6)
Initiation of treatment
Inoculation

The effect of treatment (i.p.) with ECO-04601 on the growth of newly implanted C6 tumors was evaluated using similar procedures. Statistical analysis of the mean values was done using Anova's one-way analysis of variance.

All treatments with ECO-04601 significantly decreased the growth of C6 tumors as compared to the saline-treated group.

In vivo toxicity data

Acute treament:

Table 2: Maximum tolerated dose of ECO-04601 in mice

	MTD	Observations
I.V.	≥10 mg/kg	• Highest dose tested • No deaths observed over 7 days
I.P.	30 mg/kg	• No deaths observed over 7 days
P.O	≥150 mg/kg	• No deaths observed over 7 days

MTD was evaluated on CD-1 mice (n=3-5) treated with increasing doses of ECO-04601 (1 to 10 mg/kg i.v.; 1 to 50 mg/kg i.p.; 25 to 200 mg/kg p.o.) and observed for 7 days.

Results from the acute treatment studies suggest bioavailability following oral and parenteral routes of administration.

Chronic treatment:

Figure 5: Effect of ECO-04601 on mouse body weight

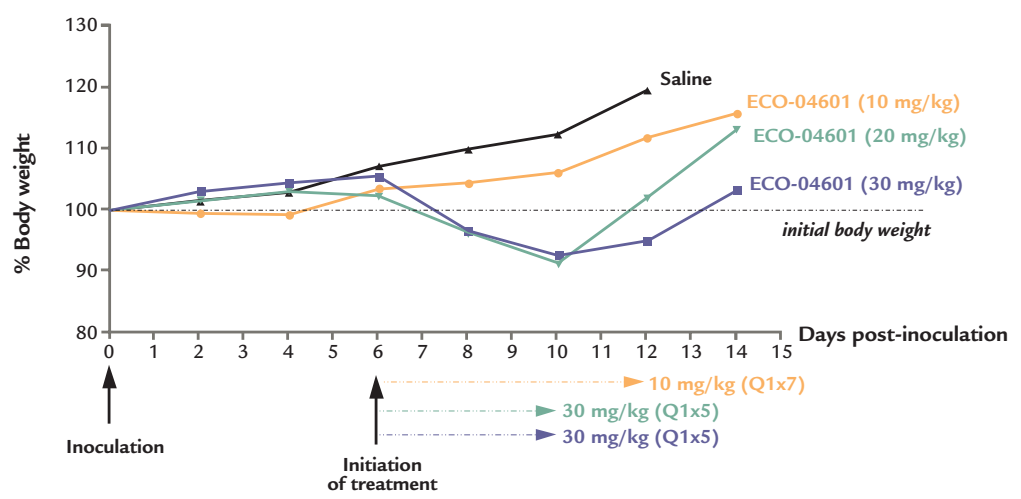
% Body weight
Saline
ECO-04601 (10 mg/kg)
ECO-04601 (20 mg/kg)
ECO-04601 (30 mg/kg)
initial body weight
Days post-inoculation
10 mg/kg (Q1x7)
30 mg/kg (Q1x5)
30 mg/kg (Q1x5)
Inoculation
Initiation of treatment

The effect of chronic administration of ECO-04601 on body weight was evaluated on C6 tumor-bearing mice. Daily i.p. treatments (10 to 30 mg/kg) started at day 6 following tumor cell inoculation. Body weight was recorded every 2 days.

Chronic administration of ECO-04601 induced a dose-dependent but reversible decrease in body weight.

CONCLUSION

- ECO-04601 has broad *in vitro* activity on different cancer cell lines, including gliomas.
- ECO-04601 inhibits the growth of glioma tumors *in vivo*.
- ECO-04601 is well-tolerated following oral and parenteral administration.
- Efficacious doses of ECO-04601 are well-tolerated in chronic treatment regimens.

Ecopia's innovative DECIPHER® technology platform allows the discovery of new anticancer agents of natural origin as demonstrated here with ECO-04601.

References:

Ahlert J, Shepard E, Lomovskaya N, Zazopoulos E, Staffa A, Bachmann BO, Huang K, Fonstein L, Czisny A, Whitwam RE, Farnet CM, Thorson JS.
The calicheamicin gene cluster and its iterative type I enediyne PKS.
Science 2002 Aug 16;297(5584):1173-6.

Zazopoulos E, Huang K, Staffa A, Liu W, Bachmann BO, Nonaka K, Ahlert J, Thorson JS, Shen B, Farnet CM.
A genomics-guided approach for discovering and expressing cryptic metabolic pathways.
Nat Biotechnol. 2003 Feb;21(2):187-90.

AACR, Orlando, FL, 27-31 March, 2004